A New Manufacturing Process to Remove Thrombogenic Factors (II, VII, IX, X, and XI) from Intravenous Immunoglobulin Gamma Preparations

Dong Hwarn Park [a], Gil Bu Kang [a], Dae Eun Kang [a], Jeung Woon Hong [a], Min Gyu Lee [b], Ki Yong Kim [a], Jeung Whan Han [b]
[a] Green Cross Corp., Ihyeon-ro 30 beon-gil, Giheung-gu, Yongin-si, Gyeonggi-do, Republic of Korea
[b] Research Center for Epigenome Regulation, School of Pharmacy, Sungkyunkwan University, Suwon 16419, Republic of Korea

INTRODUCTION

- The risk of thrombi formation and thromboembolic events is increased due to high concentrations of coagulation factors II, VII, IX, X, and particularly XIa remaining in intravenous immunoglobulin (IVIG) preparations as a result of imperfect removal during the manufacturing process.[1,2]
- IVIG preparation via ethanol precipitation, a process developed in the 1940s, effectively removes coagulation factors II, VII, IX, and X.[3,4]
- The presence of factor XIa (FXIa), however, still poses a risk of activating the coagulation cascade in the blood of patients receiving IVIG.
- It is difficult to separate FXIa from IgG via ethanol precipitation alone due to the high isoelectric point (pI 8.9-9.1) of FXIa.[5]
- An additional purification process is therefore required to remove residual FXIa in IVIG preparations.

STUDY OBJECTIVES

- To develop a new process for reducing the content of FXIa in IVIG preparations to levels below detectable limits
- To evaluate the robustness of FXIa removal in the new manufacturing process via a spiking study

COAGULATION CASCADE

Figure 1. The Coagulation Cascade

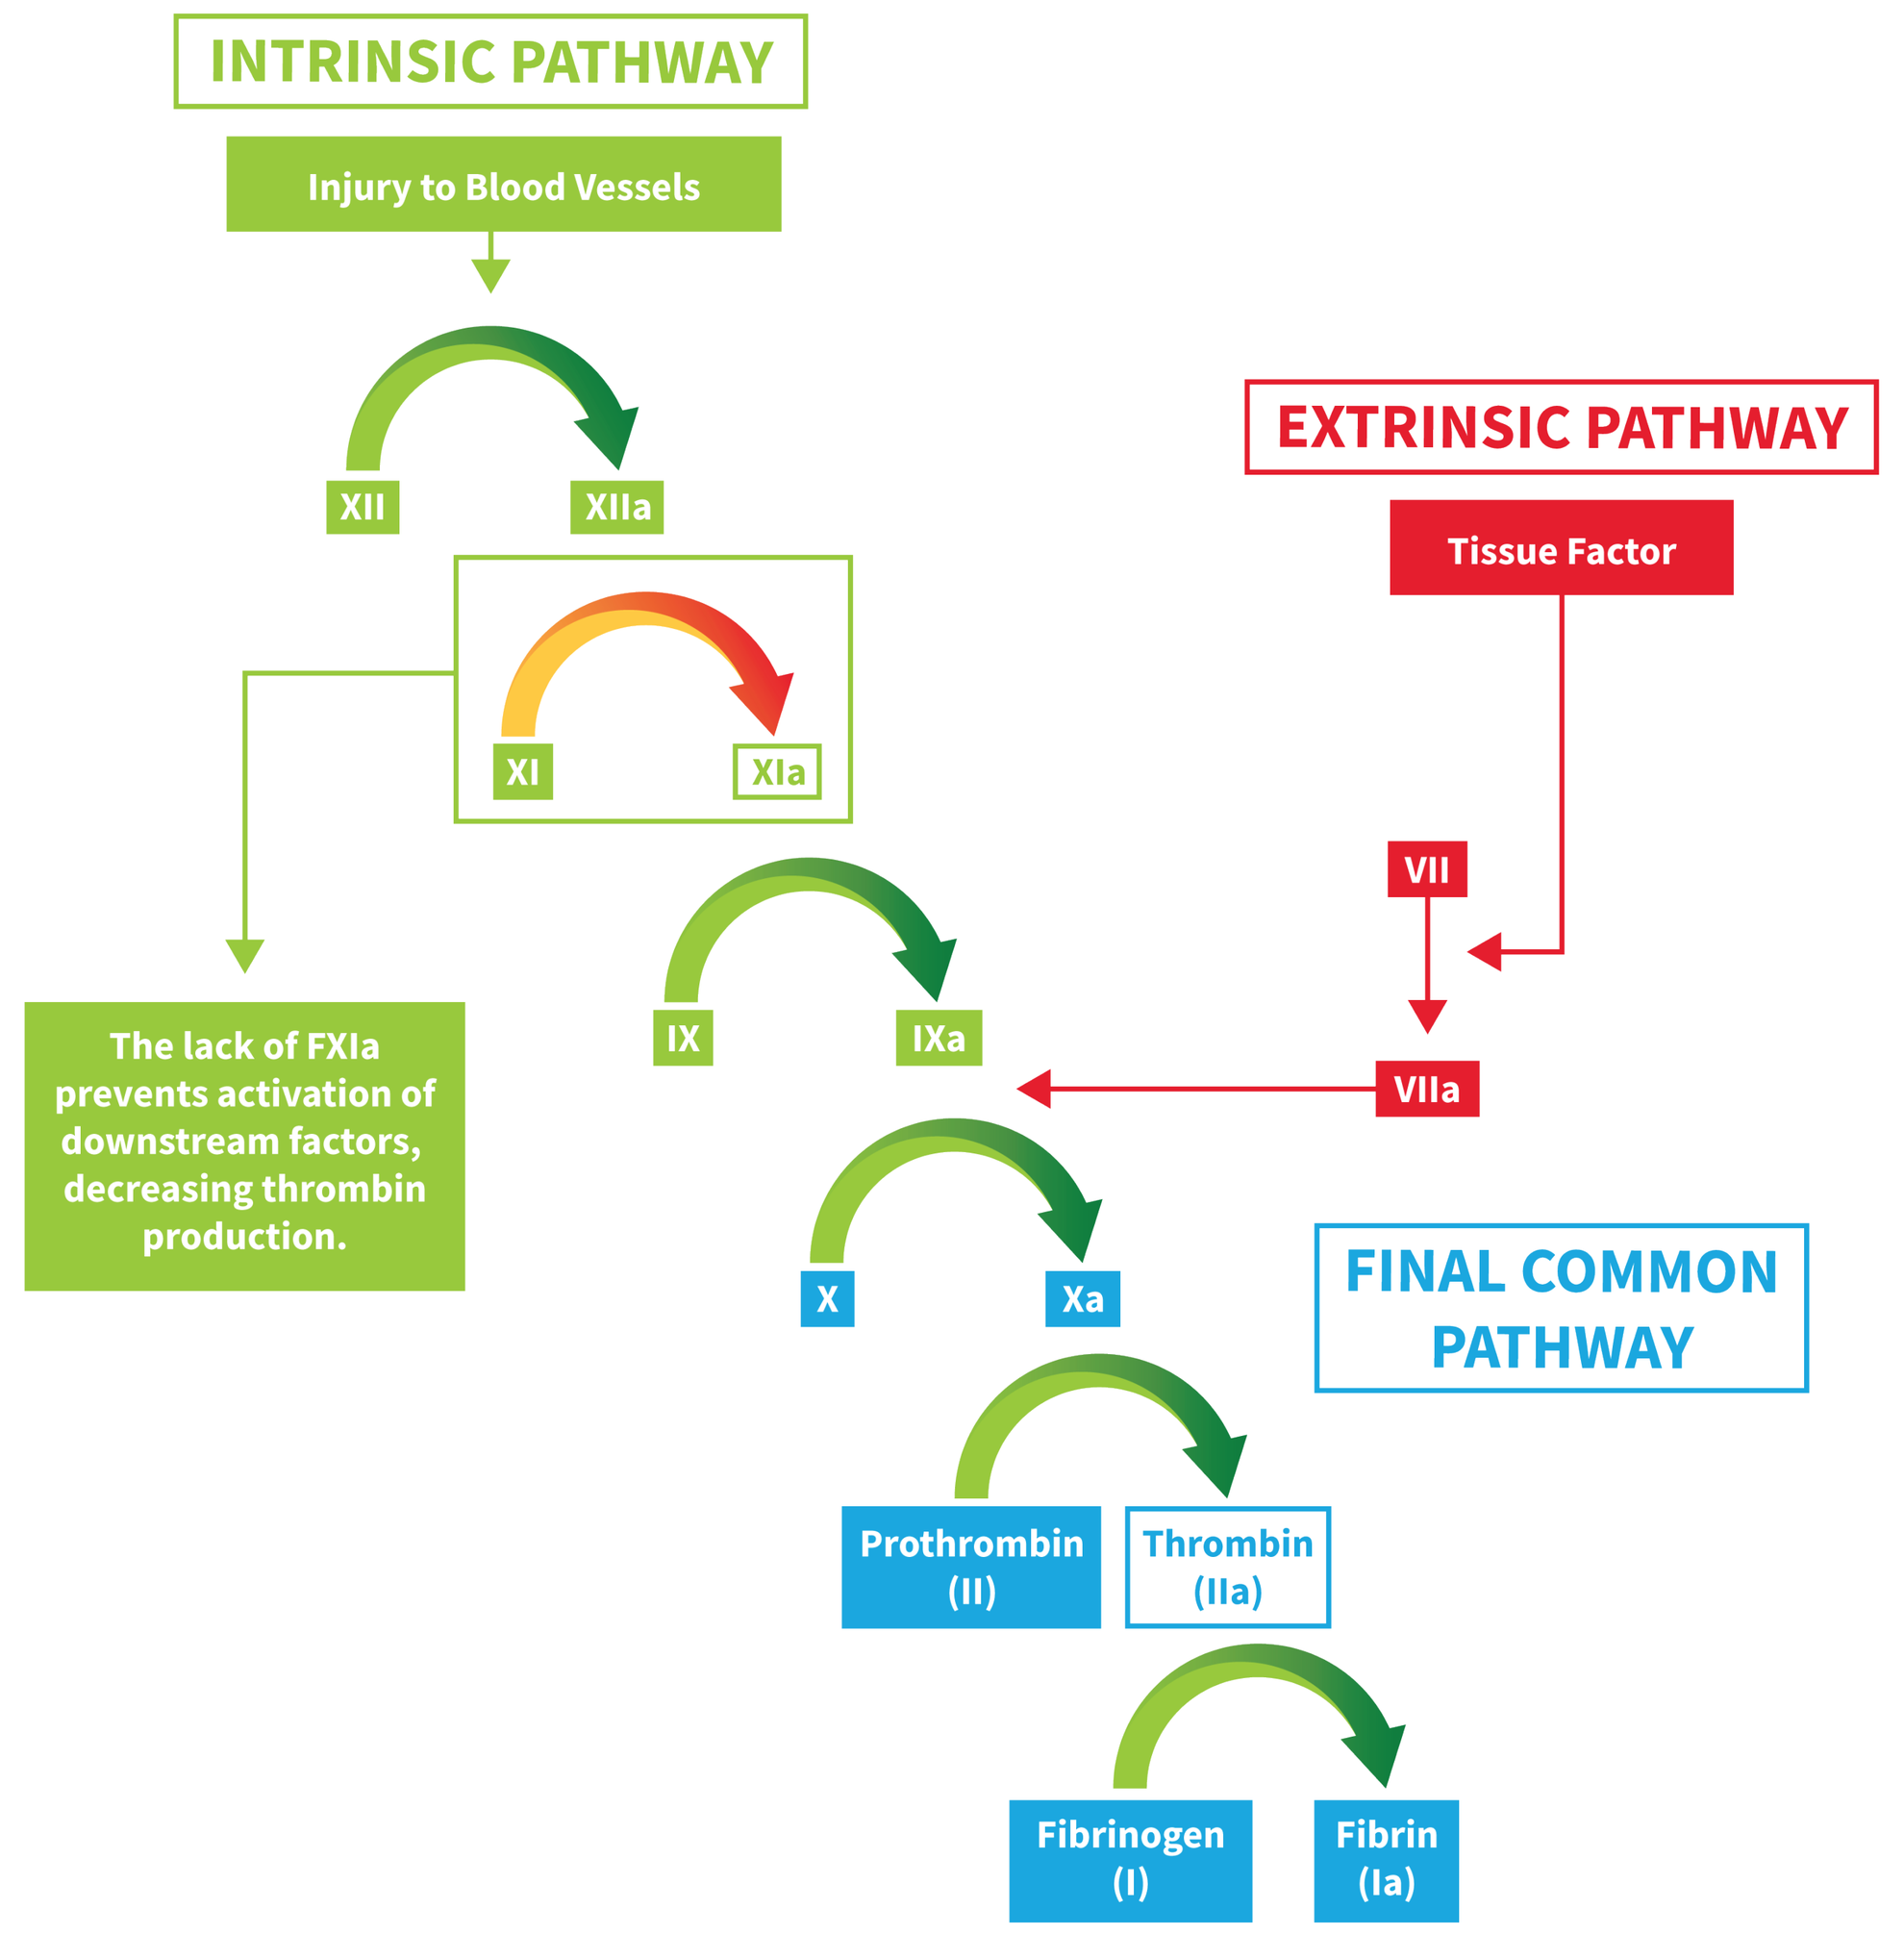
INTRINSIC PATHWAY
Injury to Blood Vessels
XII
XIIa
XI
XIa
The lack of FXIa prevents activation of downstream factors, decreasing thrombin production.
EXTRINSIC PATHWAY
Tissue Factor
VII
VIIa
IX
IXa
X
Xa
FINAL COMMON PATHWAY
Prothrombin (II)
Thrombin (IIa)
Fibrinogen (I)
Fibrin (Ia)

IVIG PURIFICATION PROCESS AND QUANTITATION OF FXIa

Figure 2. Flow Chart of Manufacturing Process for GC5101B

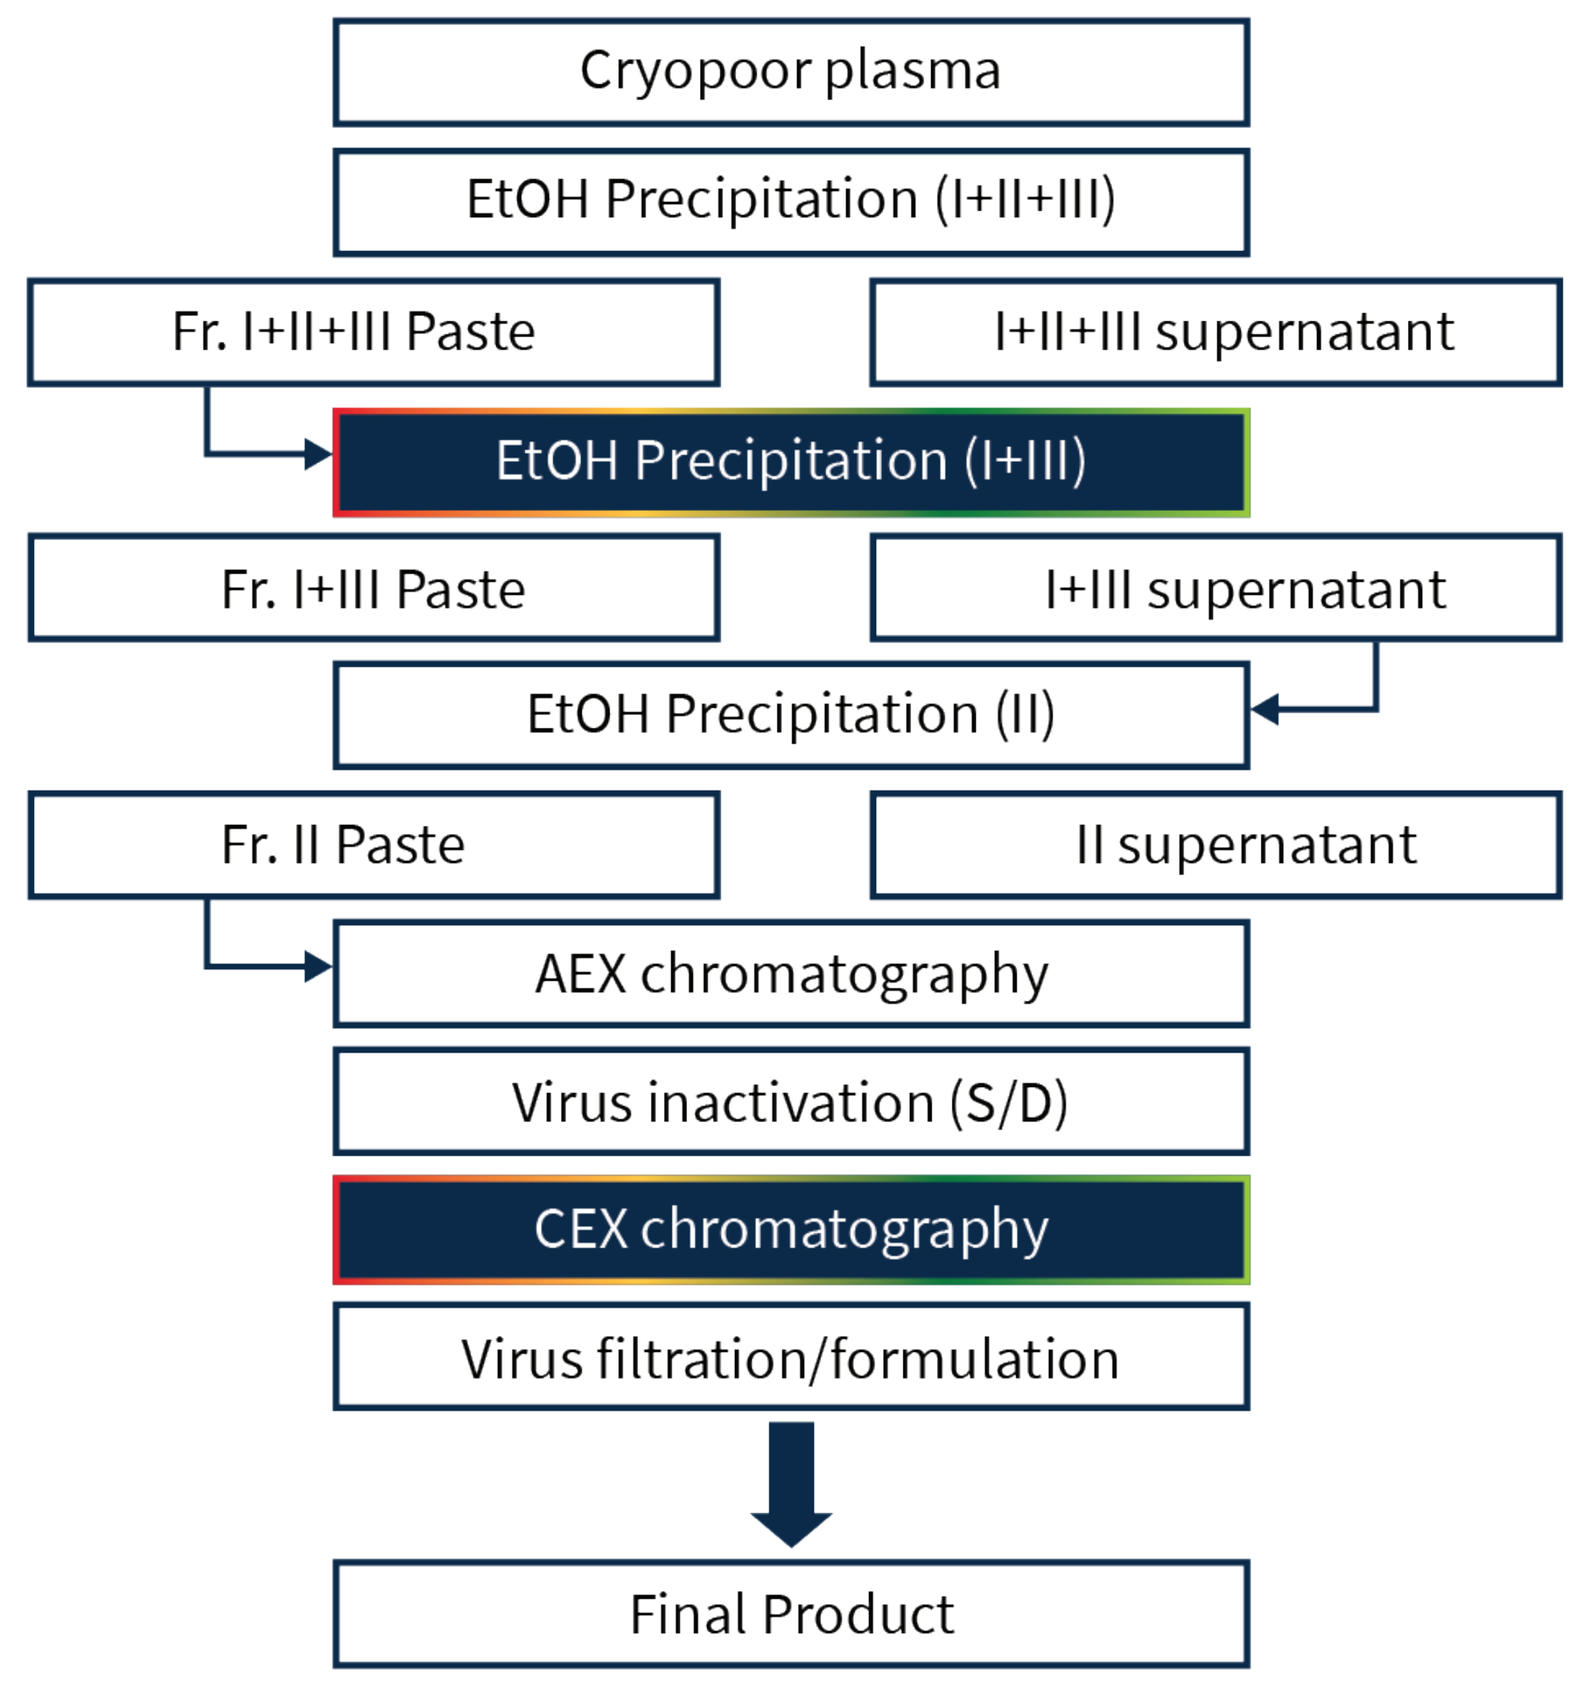
Cryopoor plasma
EtOH Precipitation (I+II+III)
Fr. I+II+III Paste
I+II+III supernatant
EtOH Precipitation (I+III)
Fr. I+III Paste
I+III supernatant
EtOH Precipitation (II)
Fr. II Paste
II supernatant
AEX chromatography
Virus inactivation (S/D)
CEX chromatography
Virus filtration/formulation
Final Product

The chromatographic cation exchange (CEX) process utilizes a new ceramic-based resin that binds IgG with 1.5 times higher capacity compared to commonly used resins in protein purification.

Procoagulant activity of the intermediate and final (GC5101B) products was evaluated using:

- Thrombin generation assay (TGA)
- Chromogenic FXIa assay
- Non-activated partial thromboplastin time (NaPTT)
- FXI/FXIa ELISA
- Western blot

Adjusting the pH of the elution buffer to 4.5 and conducting ultra/diafiltration resulted in maximal inhibition of IgG polymerization – thus establishing a stable manufacturing process for GC5101B.

RESULTS

Table 1. Results of Spiking Study

Evaluation of the reduction of FXIa contents and activity by the spiking test in cation exchange chromatography (CM Hyper D).

Process step	Pre-CEX w/o spiked		Pre-CEX w/ spiked[d]			
	ELISA[a] (µg/L)	TGA[b] (IU/L)	ELISA (µg/L)	TGA (IU/L)	ELISA (µg/L)	TGA (IU/L)
Pre-CEX	123.6	42.2	4020.1	72837.4	21002.2	447861.4
Post-CEX	<0.31	<1.56	<0.31	<1.56	2.93	<1.56
IgG recovery (%)	97.1		95.3		99.4	
Residual ratio (%)	N.C.[c]	N.C.	N.C.	N.C.	0.01	N.C.

32.5X more FXIa than normal plasma → FXIa reduced to below detection limits

170X more FXIa than normal plasma → FXIa reduced to very low levels

All samples were measured at least in duplicate.
[a] ELISA: Enzyme-linked immunosorbent assay
[b] TGA: Thrombin generation assay
[c] N.C.: Not calculated
[d] These concentrations are 32.5 times and 170 times higher than those in normal specimens.

RESULTS

Procoagulant activities were reduced to low levels as determined by:

- **Thrombin Generation Assay (TGA)**
 < 1.56 mIU/mL
- **Chromogenic FXIa Assay**
 < 0.16 mIU/mL
- **Non-activated Partial Thromboplastin Time (NaPTT)**
 > 250 seconds
- **FXI/FXIa ELISA**
 < 0.31 ng/mL

Figure 3. FXIa Remaining after Cold Ethanol Fractionation

FXIa content for each process was calculated using the step residual ratio.

1. Cryo-poor plasma (statistical results of 3 batches, mean ± SD): 100%
2. Fraction I + II + III paste: 74.4 ± 3.7
3. Fraction I + III supernatant: 5.8 ± 2.7
4. Fraction II paste: 5.2 ± 2.1. Results from 3 consecutive batches are shown; all samples measured at least in duplicate.

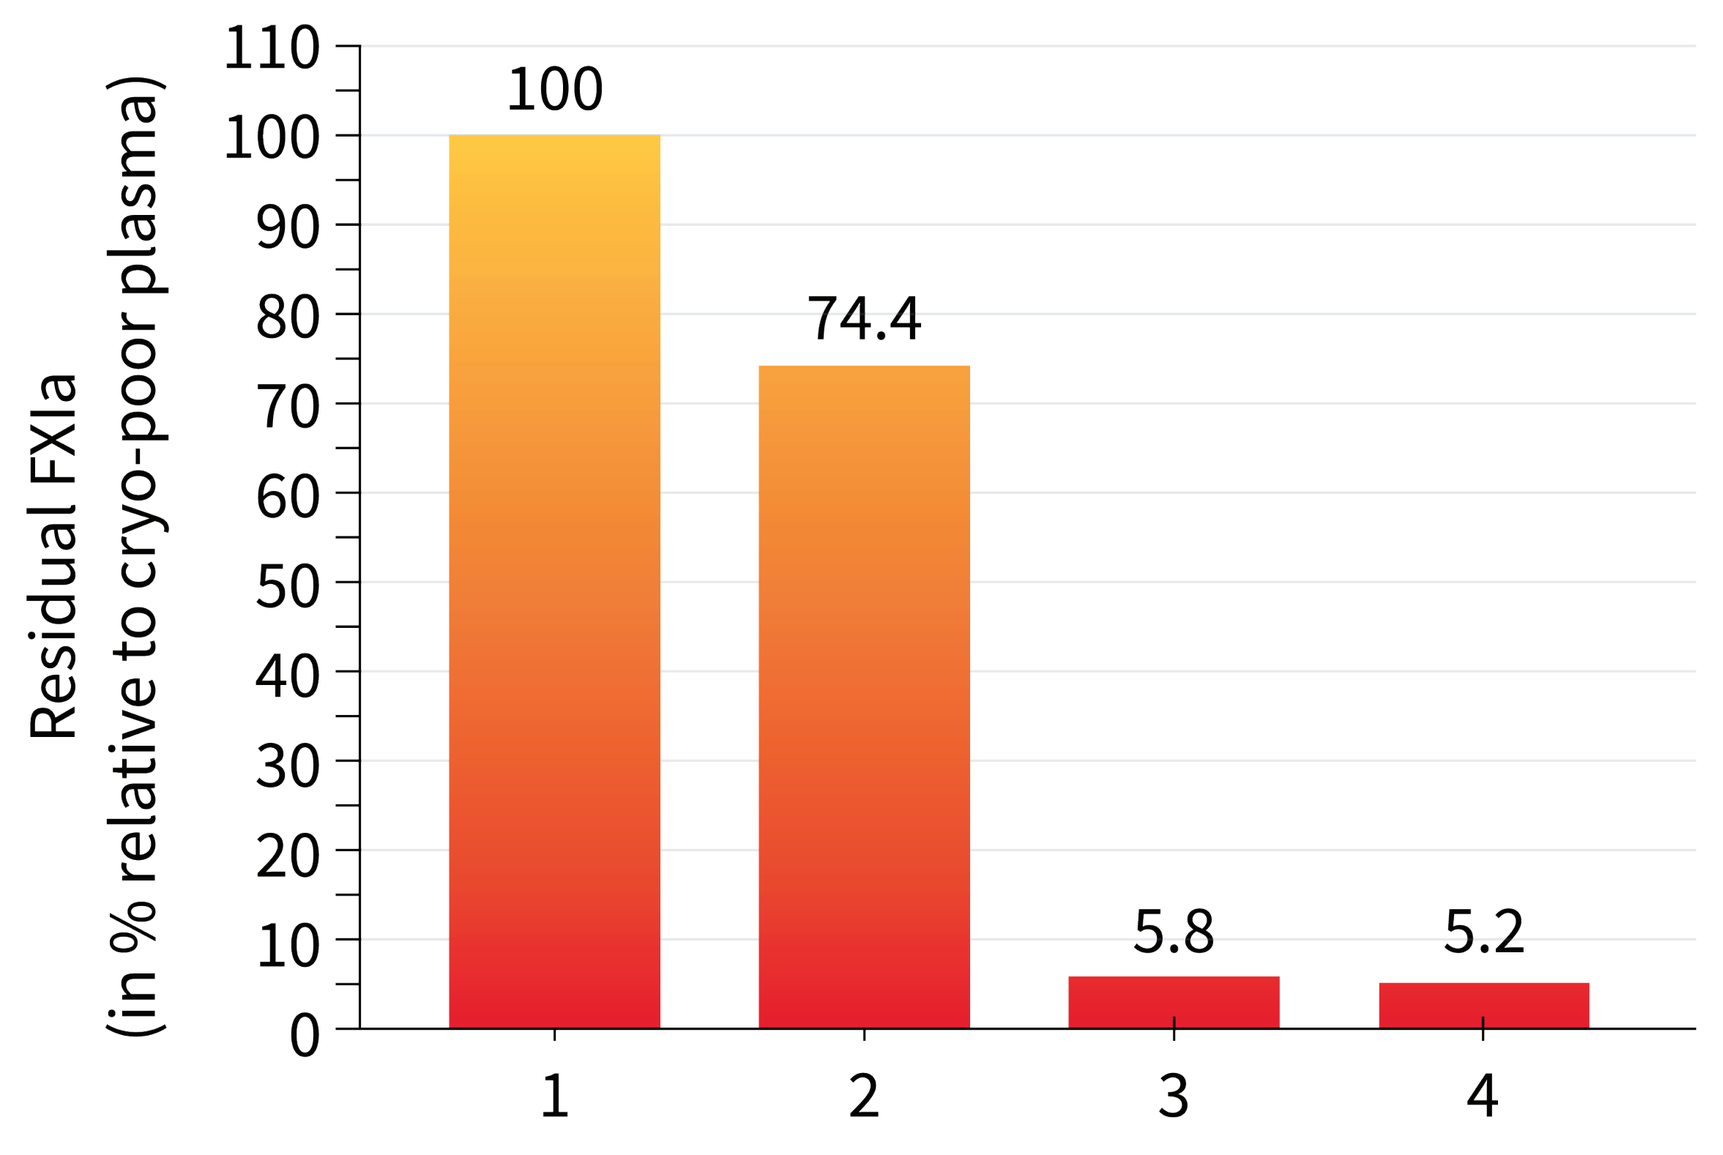
Residual FXIa
(in % relative to cryo-poor plasma)
0
10
20
30
40
50
60
70
80
90
100
110
100
74.4
5.8
5.2
1
2
3
4

RESULTS

Figure 4. Removal of Coagulation Factors by Cold Ethanol Fractionation and CEX Chromatography

1. Cryo-poor plasma (219.0 ng/mL)
2. Fraction I + II + III paste (183.0 ng/mL)
3. Fraction I + III paste (14.0 ng/mL)
4. Fraction II paste (13.0 ng/mL)
5. Pre-CEX chromatography (10.3 ng/mL)
6. Post-CEX chromatography (not detected)

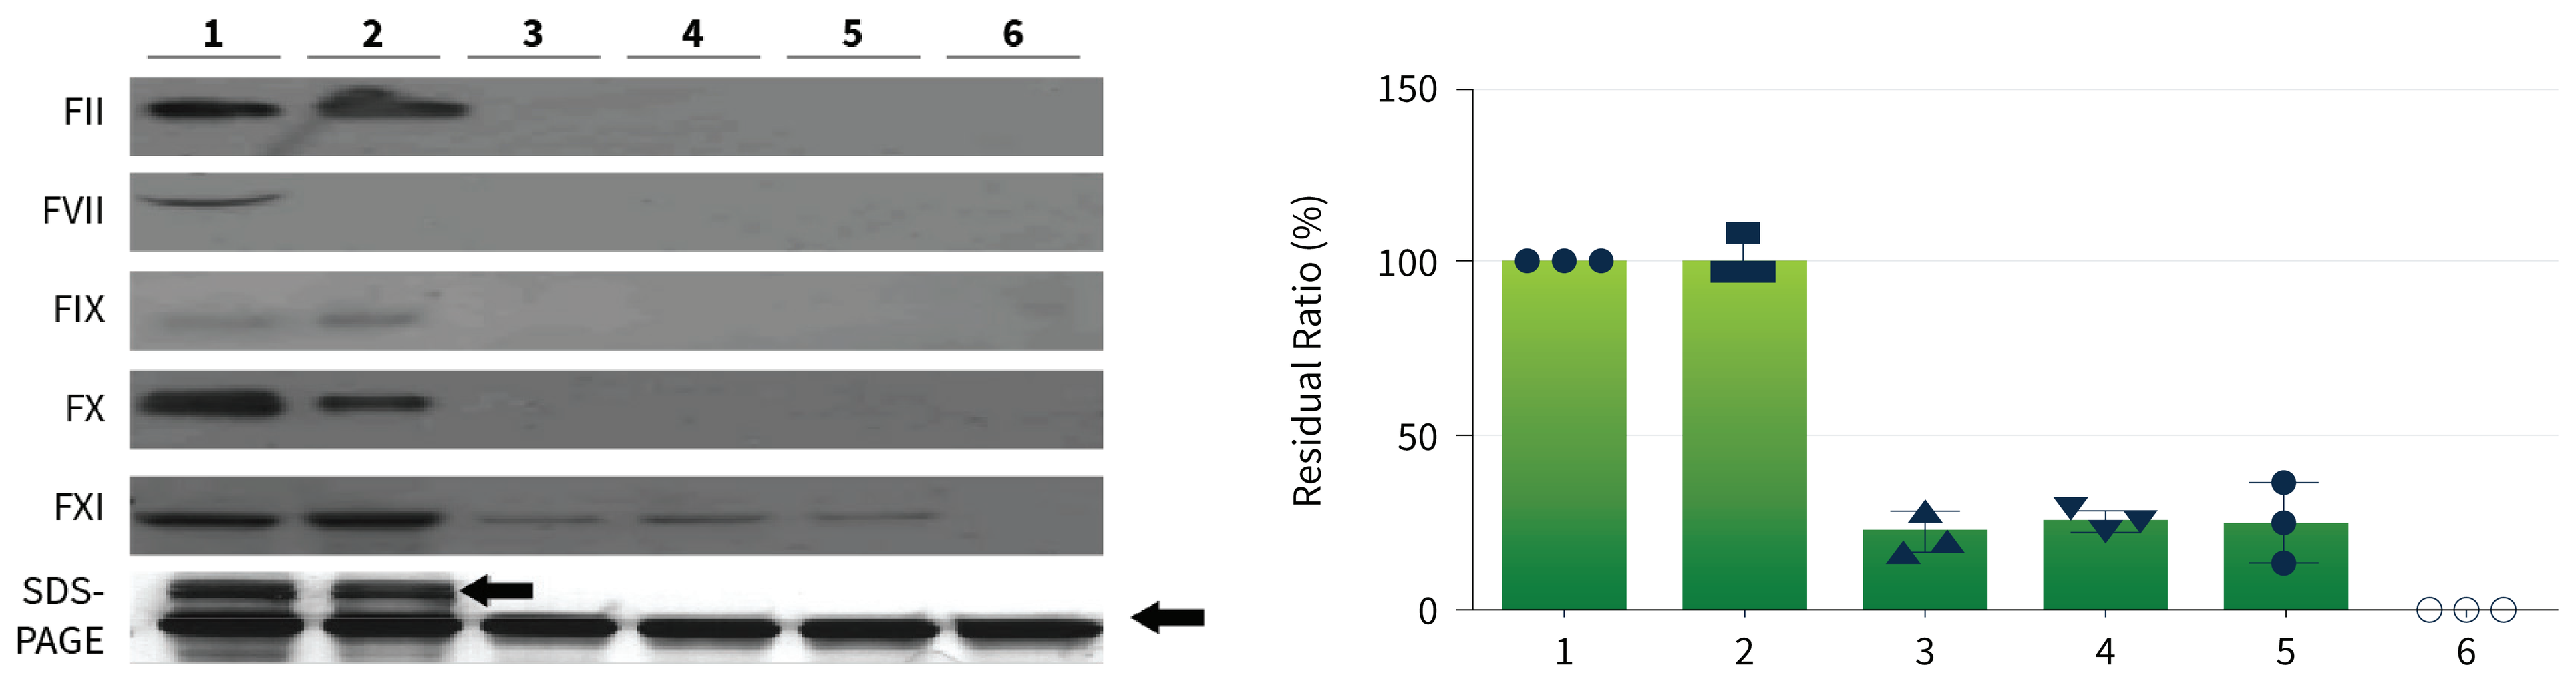
1
2
3
4
5
6
FII
FVII
FIX
FX
FXI
SDS-PAGE
Residual Ratio (%)
150
100
50
0
1
2
3
4
5
6

A. Image of the western blot with control (non-reduced condition, 4-20% Tris-glycine gel, upper arrow: IgG monomer, 150 kDa; lower arrow: IgG polymer)
Loading amount is 5.0 mg of IgG and concentration of FXI/FXIa calculated by ELISA

B. Band density of FXI/FXIa calculated using Prizm ver. 6.0

DISCUSSION

- Coagulation factors II, VII, IX, and X were removed via cold ethanol precipitation and FXIa was successfully removed to undetectable levels using CEX chromatography.
- The robustness of this process was evaluated through a spiking study. With the spiking assay, we showed that the high binding capacity resin (> 100.0 g/L) further increased the production efficiency.
- The spiking study including cation exchange chromatography was conducted using 32.5 and 170 times the concentration of FXIa as that present in normal specimens, and the results were analyzed using ELISA and TGA.
- The results demonstrate the ability of CEX chromatography to effectively remove procoagulant activities to below the detection limit (except by NaPTT).
- A reduced risk of thromboembolic events may result from this new IVIG fractionation process.

CONCLUSION

- The use of our manufacturing process may improve the safety of IVIG by reducing FXIa levels to below the detection limit, potentially minimizing the risk of thromboembolic events associated with the use of IVIG products.
- Our results are difficult to directly compare with previous studies, as we have used different test and standard materials to quantitate FXIa activity.
- Additional in vivo assays (such as the Wessler test) would be necessary to demonstrate the safety of the preparations more precisely.

REFERENCES

1. Roemisch JR, Kaar W, Zoechling A, et al. WebmedCentral *Immunotherapy.* 2011;2:WMC002002.
2. Wolberg AS, Kon RH, Monroe DM, Hoffman M. *Am J Hematol.* 2000;65:30-34.
3. Cohn EJ, Strong LE, Hughes Jr WL, et al. *J Am Chem Soc.* 1946;68:459-75.
4. Oncley JL, Melin M, Richert DA, et al. *J Am Chem Soc.* 1949;71:541-50.
5. Burnouf-Radosevich M, Burnouf T. *Transfusion.* 1992;32:861-7.

GC